USE OF A **NOVEL LOW – FAT HYDROLYSED DIET** IN DOGS WITH CHRONIC ENTEROPATHY

Claudia Hernández | Luis Feo | Jordi Puig
Internal Medicine Department. AniCura Ars Veterinary Hospital. Barcelona, Spain.

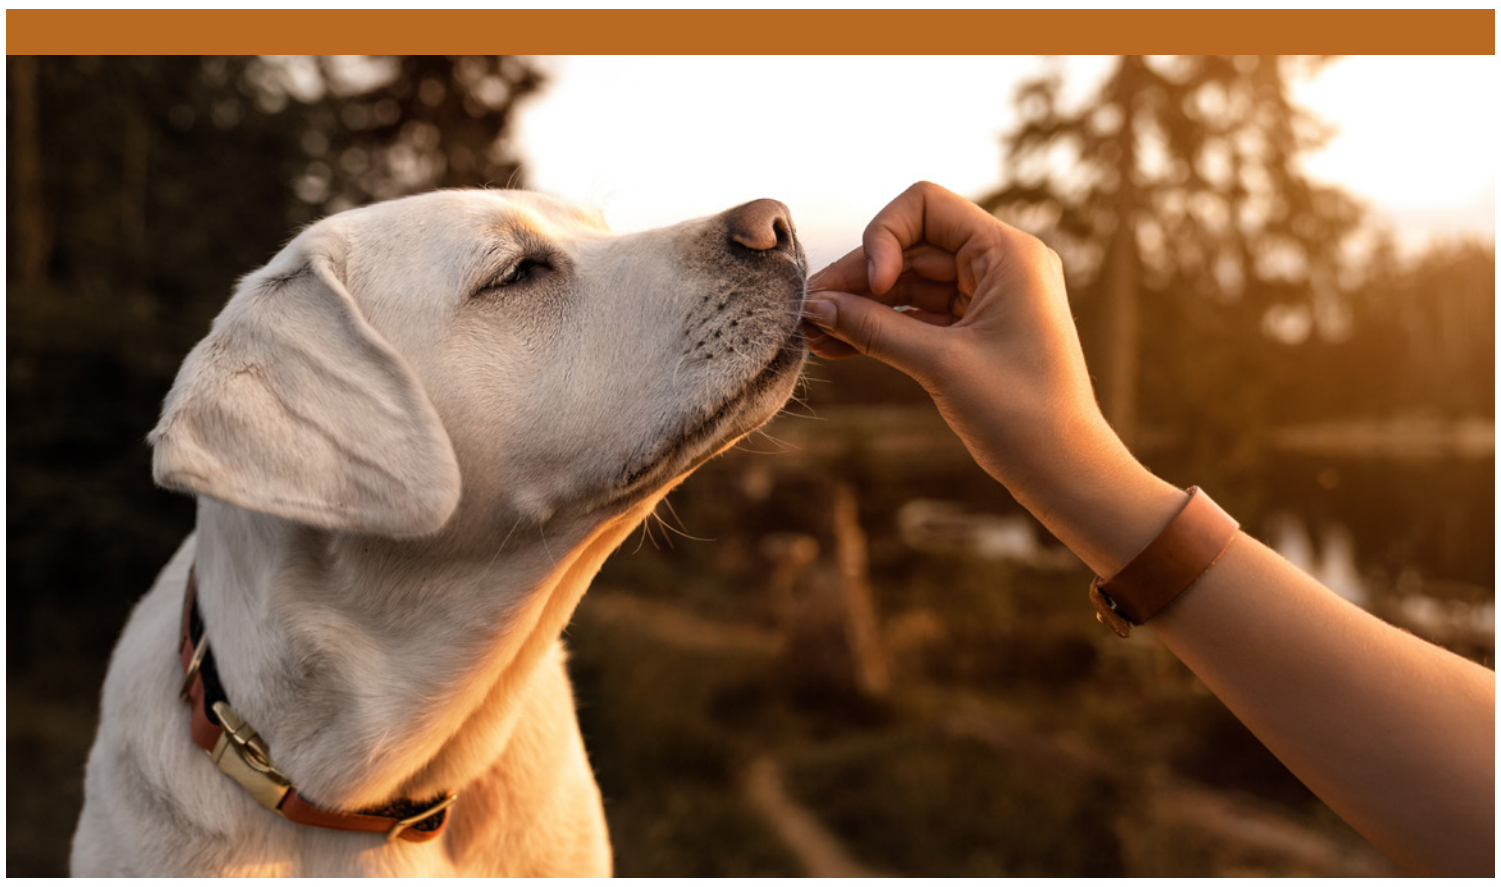

INTRODUCTION

The role of diet is paramount in both the development and management of chronic enteropathies (CE). It is through dietary interventions such as novel or hydrolysed proteins, low-fat content, high fibre or increased digestibility that effective management can be achieved.

Hydrolysed diets are pivotal in managing dogs with CE due to their demonstrated efficacy.[1-3] Gastrointestinal (GI) diseases in dogs may increase colonic fat transit caused by lowered digestion and absorption, leading to dysbiosis, epithelial cell impairment, and increased fluid secretion into the colon.[2] Dietary fat restriction is used as a strategy in managing GI motility disorders, lymphangiectasia or pancreatitis. Long–chain triglycerides from the diet, transported into the lymphatic vessels, may result in swelling and inflammation of the lymphatics, leading to enhanced protein leakage in dogs with primary or secondary lymphangiectasia. Fat-rich diets may promote slower gastric emptying and lead to deterioration in clinical signs such as vomiting or regurgitation.[3]

Recently, a commercial diet that combines the two aforementioned dietary strategies has been available for clinical use in canine patients. This study aimed to investigate using a novel, low-fat, hydrolysed diet to manage dogs with chronic enteropathy.

MATERIALS & METHODS

Study Design- Prospective cohort study.

Patients - Client-owned dogs diagnosed with CE (persistant chronic GI signs lasting longer than three weeks or a 6-month history of intermittent GI signs with the exclusion of specific infectious, structural and metabolic cuases of chronic GI signs).

Group A
Patients who have never tried hydrolysed dietary trials or novel protein diets before.

A | B

Group B
Patients who underwent dietary trials featuring hydrolysed commercial diets or novel protein diets

Diet - The study's test diet was the dry formulation of SPECIFIC Digestive Support Low Fat CID-LF provided by Dechra Veterinary Products SL. The study diet was fed exclusively. It has a fat-restricted profile (2,01g of fat/100 kcal of metabolisable energy) and is based on hydrolysed salmon protein.

EVALUATION TIME - POINTS

T0 (First Visit)	**T1** (1 month follow up)	**T2** (3 month follow up)
Start diet trial		
Signalment Previous treatments/diets Clinical Signs CIBDAI Diahorrea Type	Signalment Previous treatments/diets Clinical Signs CIBDAI Diahorrea Type	Signalment Previous treatments/diets Clinical Signs CIBDAI Diahorrea Type
CBC, Biochemistry, electrolytes Additional testing	CBC, Biochemistry, electrolytes Additional testing	CBC, Biochemistry, electrolytes Additional testing
Palatability testing		

Figure 1. Clinical data were collected at three time points during the diet trial. Clinical efficacy data and palatability were assessed at each time point. CIBDAI: Canine Inflammatory Bowel Disease Activity Index).[4]

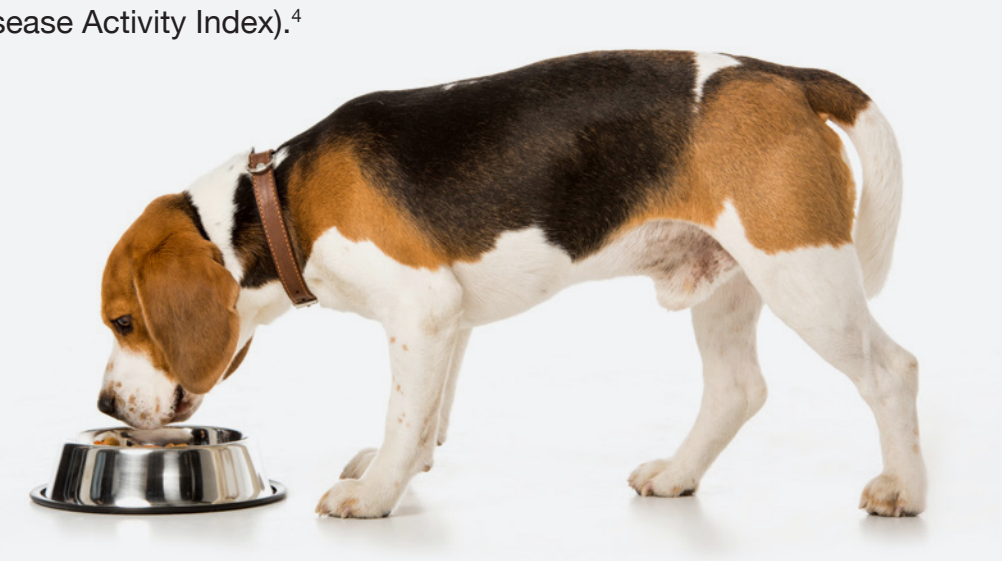

OUTCOME MEASURES

Complete response	CR	Relief of clinical signs. CIBDAI scores decrease by 50 % or a value of ≤ 3.
Partial response	PR	Improvement but persistence of GI clinical signs. CIBDAI scores decrease by 50 % or value of ≤ 3.
No response	NR	Not noticeable change in the frequency and severity of clinical signs and not meeting CR and PR criteria.

RESULTS

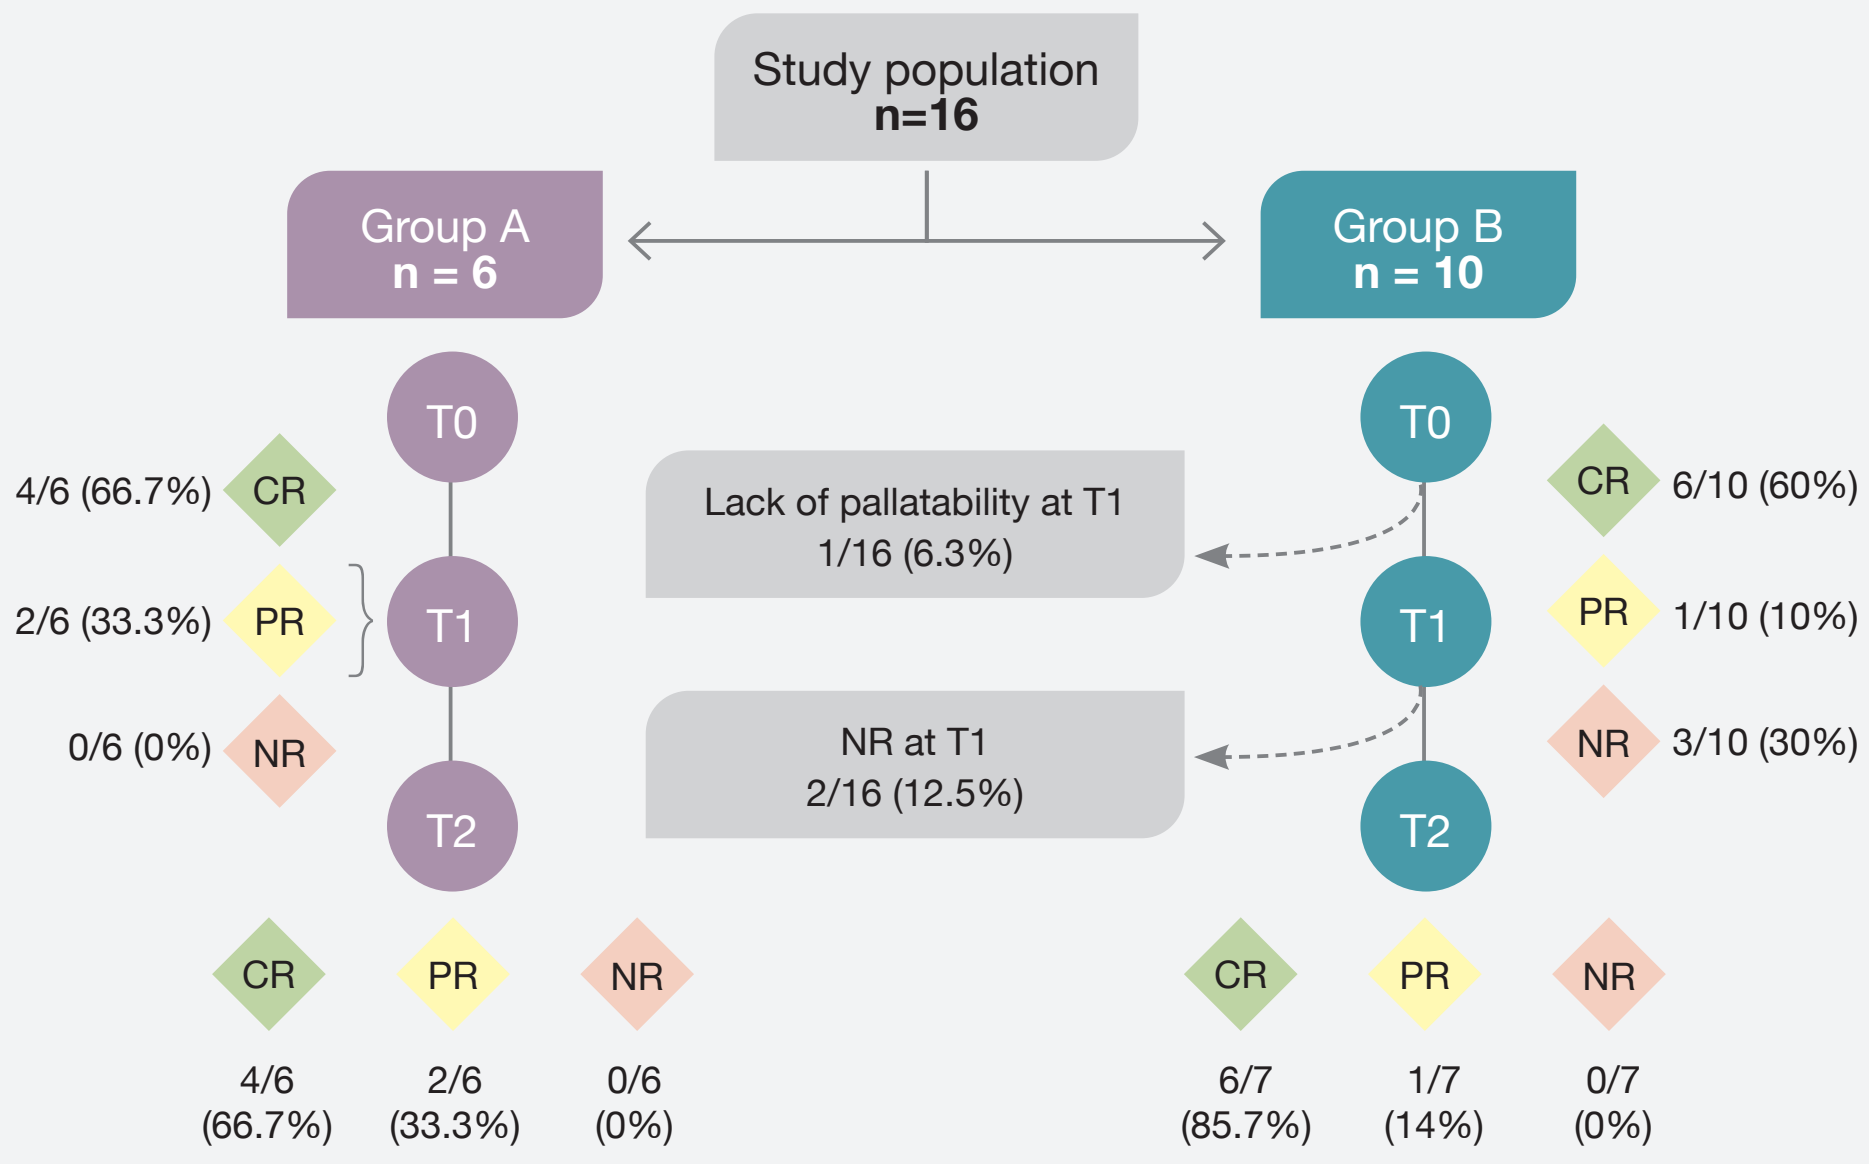
Study population
n=16
Group A
n = 6
Group B
n = 10
T0
T1
T2
4/6 (66.7%) CR
2/6 (33.3%) PR
0/6 (0%) NR
Lack of pallatability at T1
1/16 (6.3%)
NR at T1
2/16 (12.5%)
CR 6/10 (60%)
PR 1/10 (10%)
NR 3/10 (30%)
CR 4/6 (66.7%)
PR 2/6 (33.3%)
NR 0/6 (0%)
CR 6/7 (85.7%)
PR 1/7 (14%)
NR 0/7 (0%)

Figure 2. Clinical efficacy of the diet trial at the three different time – points.
CR: Complete response; ***PR:*** Partial response; **NR:** No response.

At T0, no statistically significant differences (p=0.47) in the CIBDAI score between groups A and B were observed.

- The overall CR, PR and NR in the total population were 63.6%, 18.2% and 18.2%, respectively.
- All patients who experienced a CR or PR at T1 maintained the CIBDAI improvement at T2.
- NR included 2 dogs that did not respond to diet despite adequate palatability (T1), and 1 dog who could not receive the diet due to poor palatability (T0).
- The diet was well-accepted and highly palatable to most dogs (12/16; 75%).

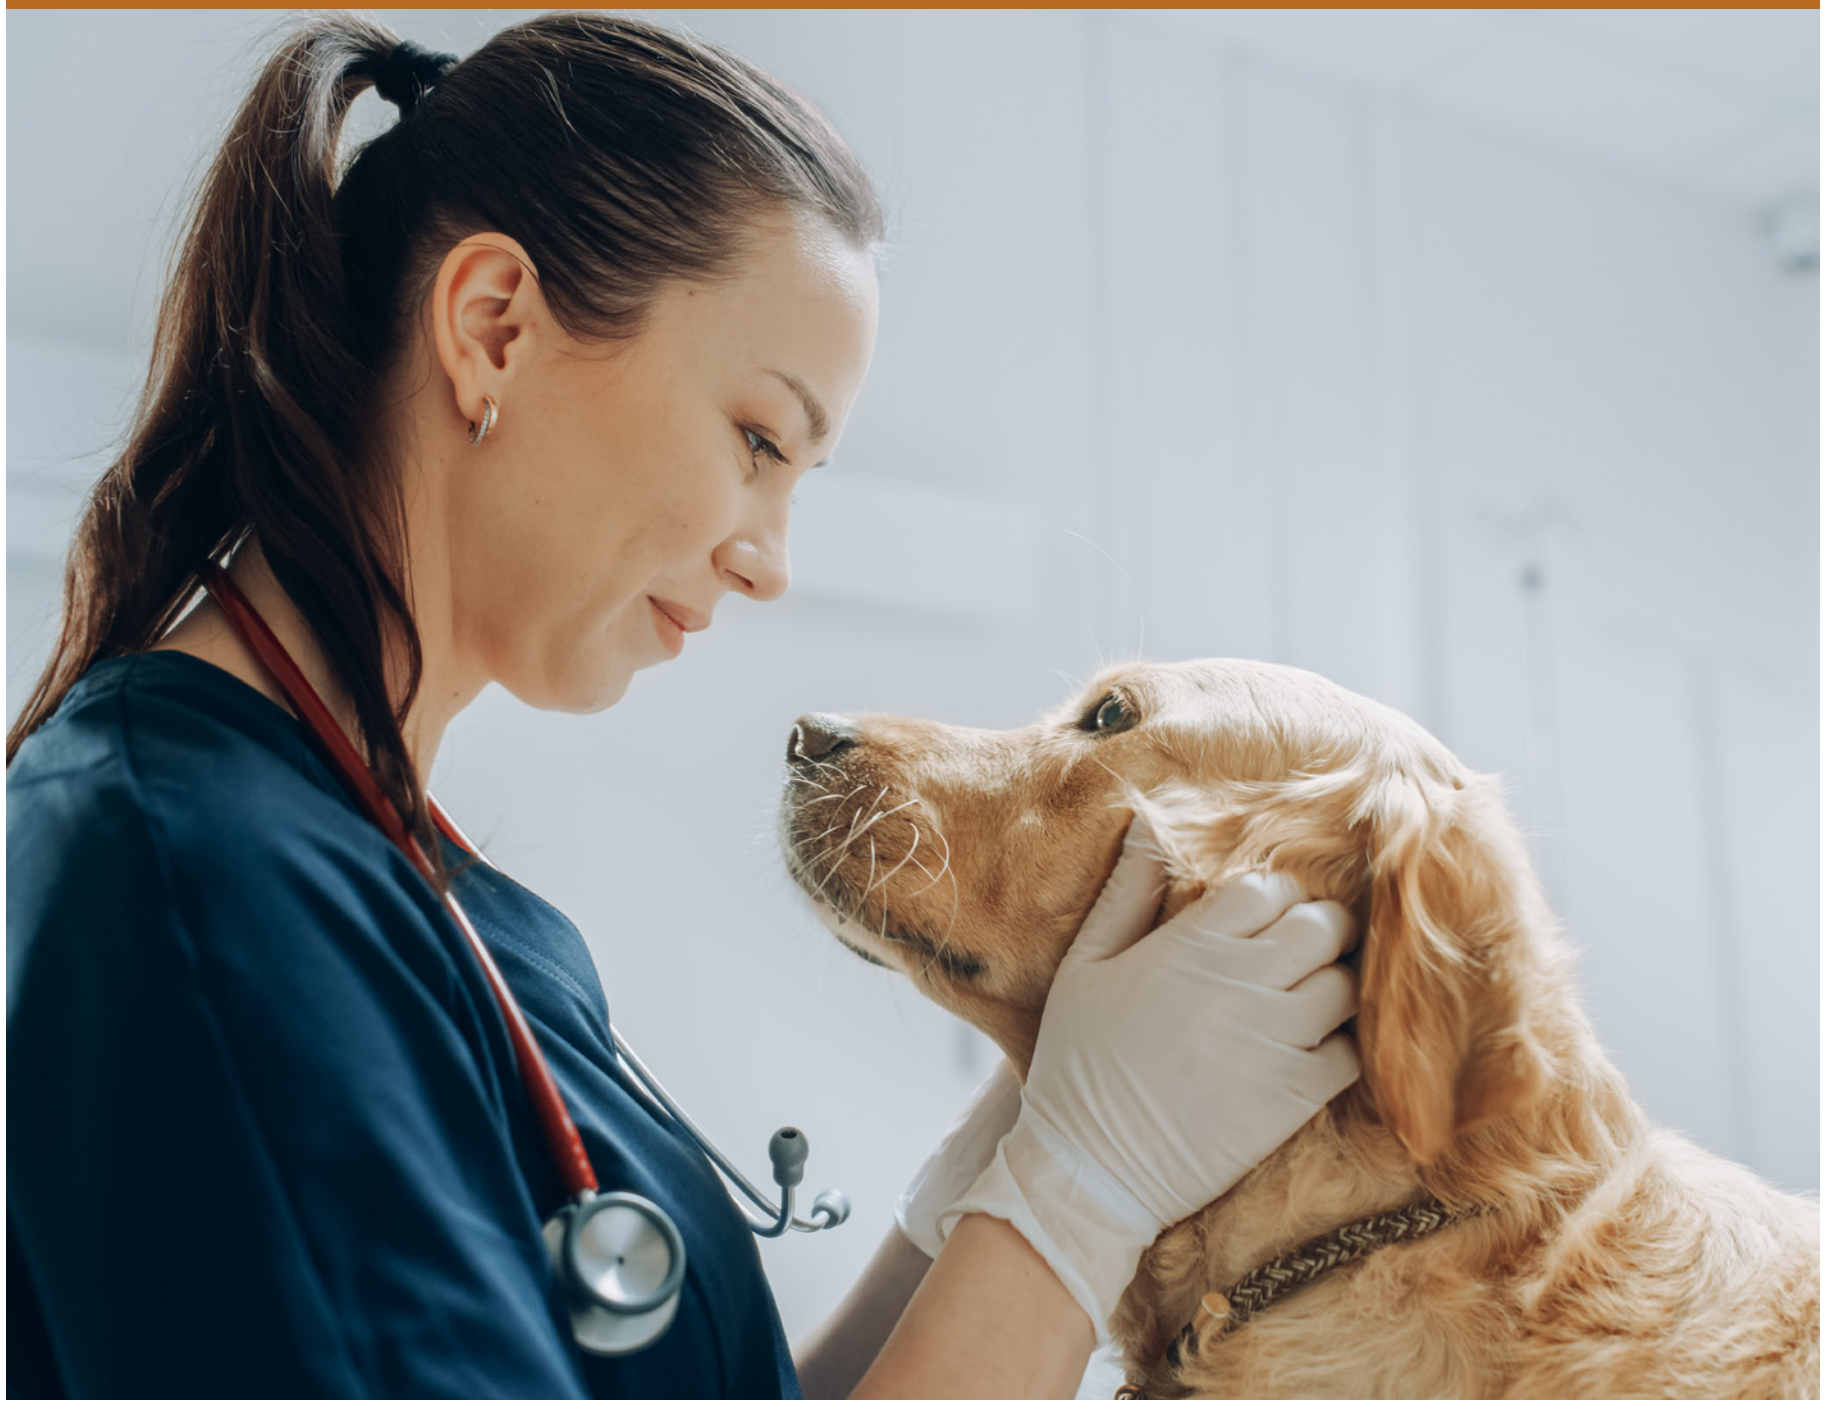

CONCLUSION

This study suggests that a low-fat, hydrolysed protein diet may be suitable for managing diet-responsive enteropathies in dogs. In this population, the diet led to an overall response of 81 % (CR and PR patients), emphasising the importance of nutrition management as a cornerstone of the therapeutic approach to chronic enteropathies.

Additionally, the high proportion of dogs who experienced a CR suggests this dietary option could be offered as an alternative when other dietary strategies have proven ineffective.

REFERENCES

1. Mandigers PJ, Biourge V, van den Ingh TS, Ankringa N, German AJ. A randomized, open-label, positively-controlled field trial of a hydrolyzed protein diet in dogs with chronic small bowel enteropathy. J Vet Intern Med. 2010 Nov-Dec;24(6):1350-7.

2. Kathrani A. Dietary and Nutritional Approaches to the Management of Chronic Enteropathy in Dogs and Cats. Vet Clin North Am Small Anim Pract. 2021 Jan;51(1):123-136.

3. Tolbert MK, Murphy M, Gaylord L, Witzel-Rollins A. Dietary management of chronic enteropathy in dogs. J Small Anim Pract. 2022 Jun;63(6):425-434.

4. Jergens AE, Schreiner CA, Frank DE, Niyo Y, Ahrens FE, Eckersall PD, Benson TJ, Evans R. A scoring index for disease activity in canine inflammatory bowel disease. J Vet Intern Med. 2003 May-Jun;17(3):291-7.

Dechra Veterinary Products | Sansaw Business Park | Hadnall | Shrewsbury | Shropshire | SY4 4AS | United Kingdom | T: +44 (0) 1939 211200 | E: info.uk@dechra.com

Dechra